Evolution of Clinical Abuse Liability Study Design for Modern Oral Nicotine Pouches

Kristen Prevette, Brian Keyser, Robert Leverette, John Darnell, Rebecca Garcia, and John W. Caraway
RAI Services Company, Winston-Salem, NC, USA

Introduction

Clinical abuse liability (AL) studies help evaluate the dependence potential of tobacco and nicotine products, typically using nicotine pharmacokinetics (PK) and subjective responses after controlled product use. Our previous modern oral nicotine pouch studies assessed these endpoints for 240 minutes after a 30-minute product use session, with usual brand (UBCC) combustible cigarettes and nicotine replacement therapy (NRT) as high and low AL comparators. Here, we describe an evolved modern oral nicotine pouch AL study design that assesses both acute and extended use and includes UBCC and moist snuff comparators. The proposed design begins with an initial 30-minute single-use phase, followed by one-hour assessments of subjective effects and PK. Then participants enter a 7-hour ad libitum phase with permitted additional product use and serial PK sampling. This approach captures nicotine uptake, liking, craving relief, and product effects under acute product use conditions. The ad libitum phase enables characterization of additional product consumption and nicotine exposure under conditions of repeated use. Including UBCC and moist snuff comparators provides AL context across combustible and smokeless tobacco product categories. Overall, the design offers a more comprehensive framework for characterizing modern oral nicotine pouch abuse liability.

Comparison of Study Designs

Endpoint	Cigarette/NRT	Moist Snuff	Proposed
Acute PK	✓	✓	✓
Acute subjective effects	✓	✓	✓
High-AL comparator	✓		✓
Oral tobacco comparator		✓	✓
Repeated product use			✓
Use behavior assessment			✓
Extended nicotine exposure			✓

Current Study Framework

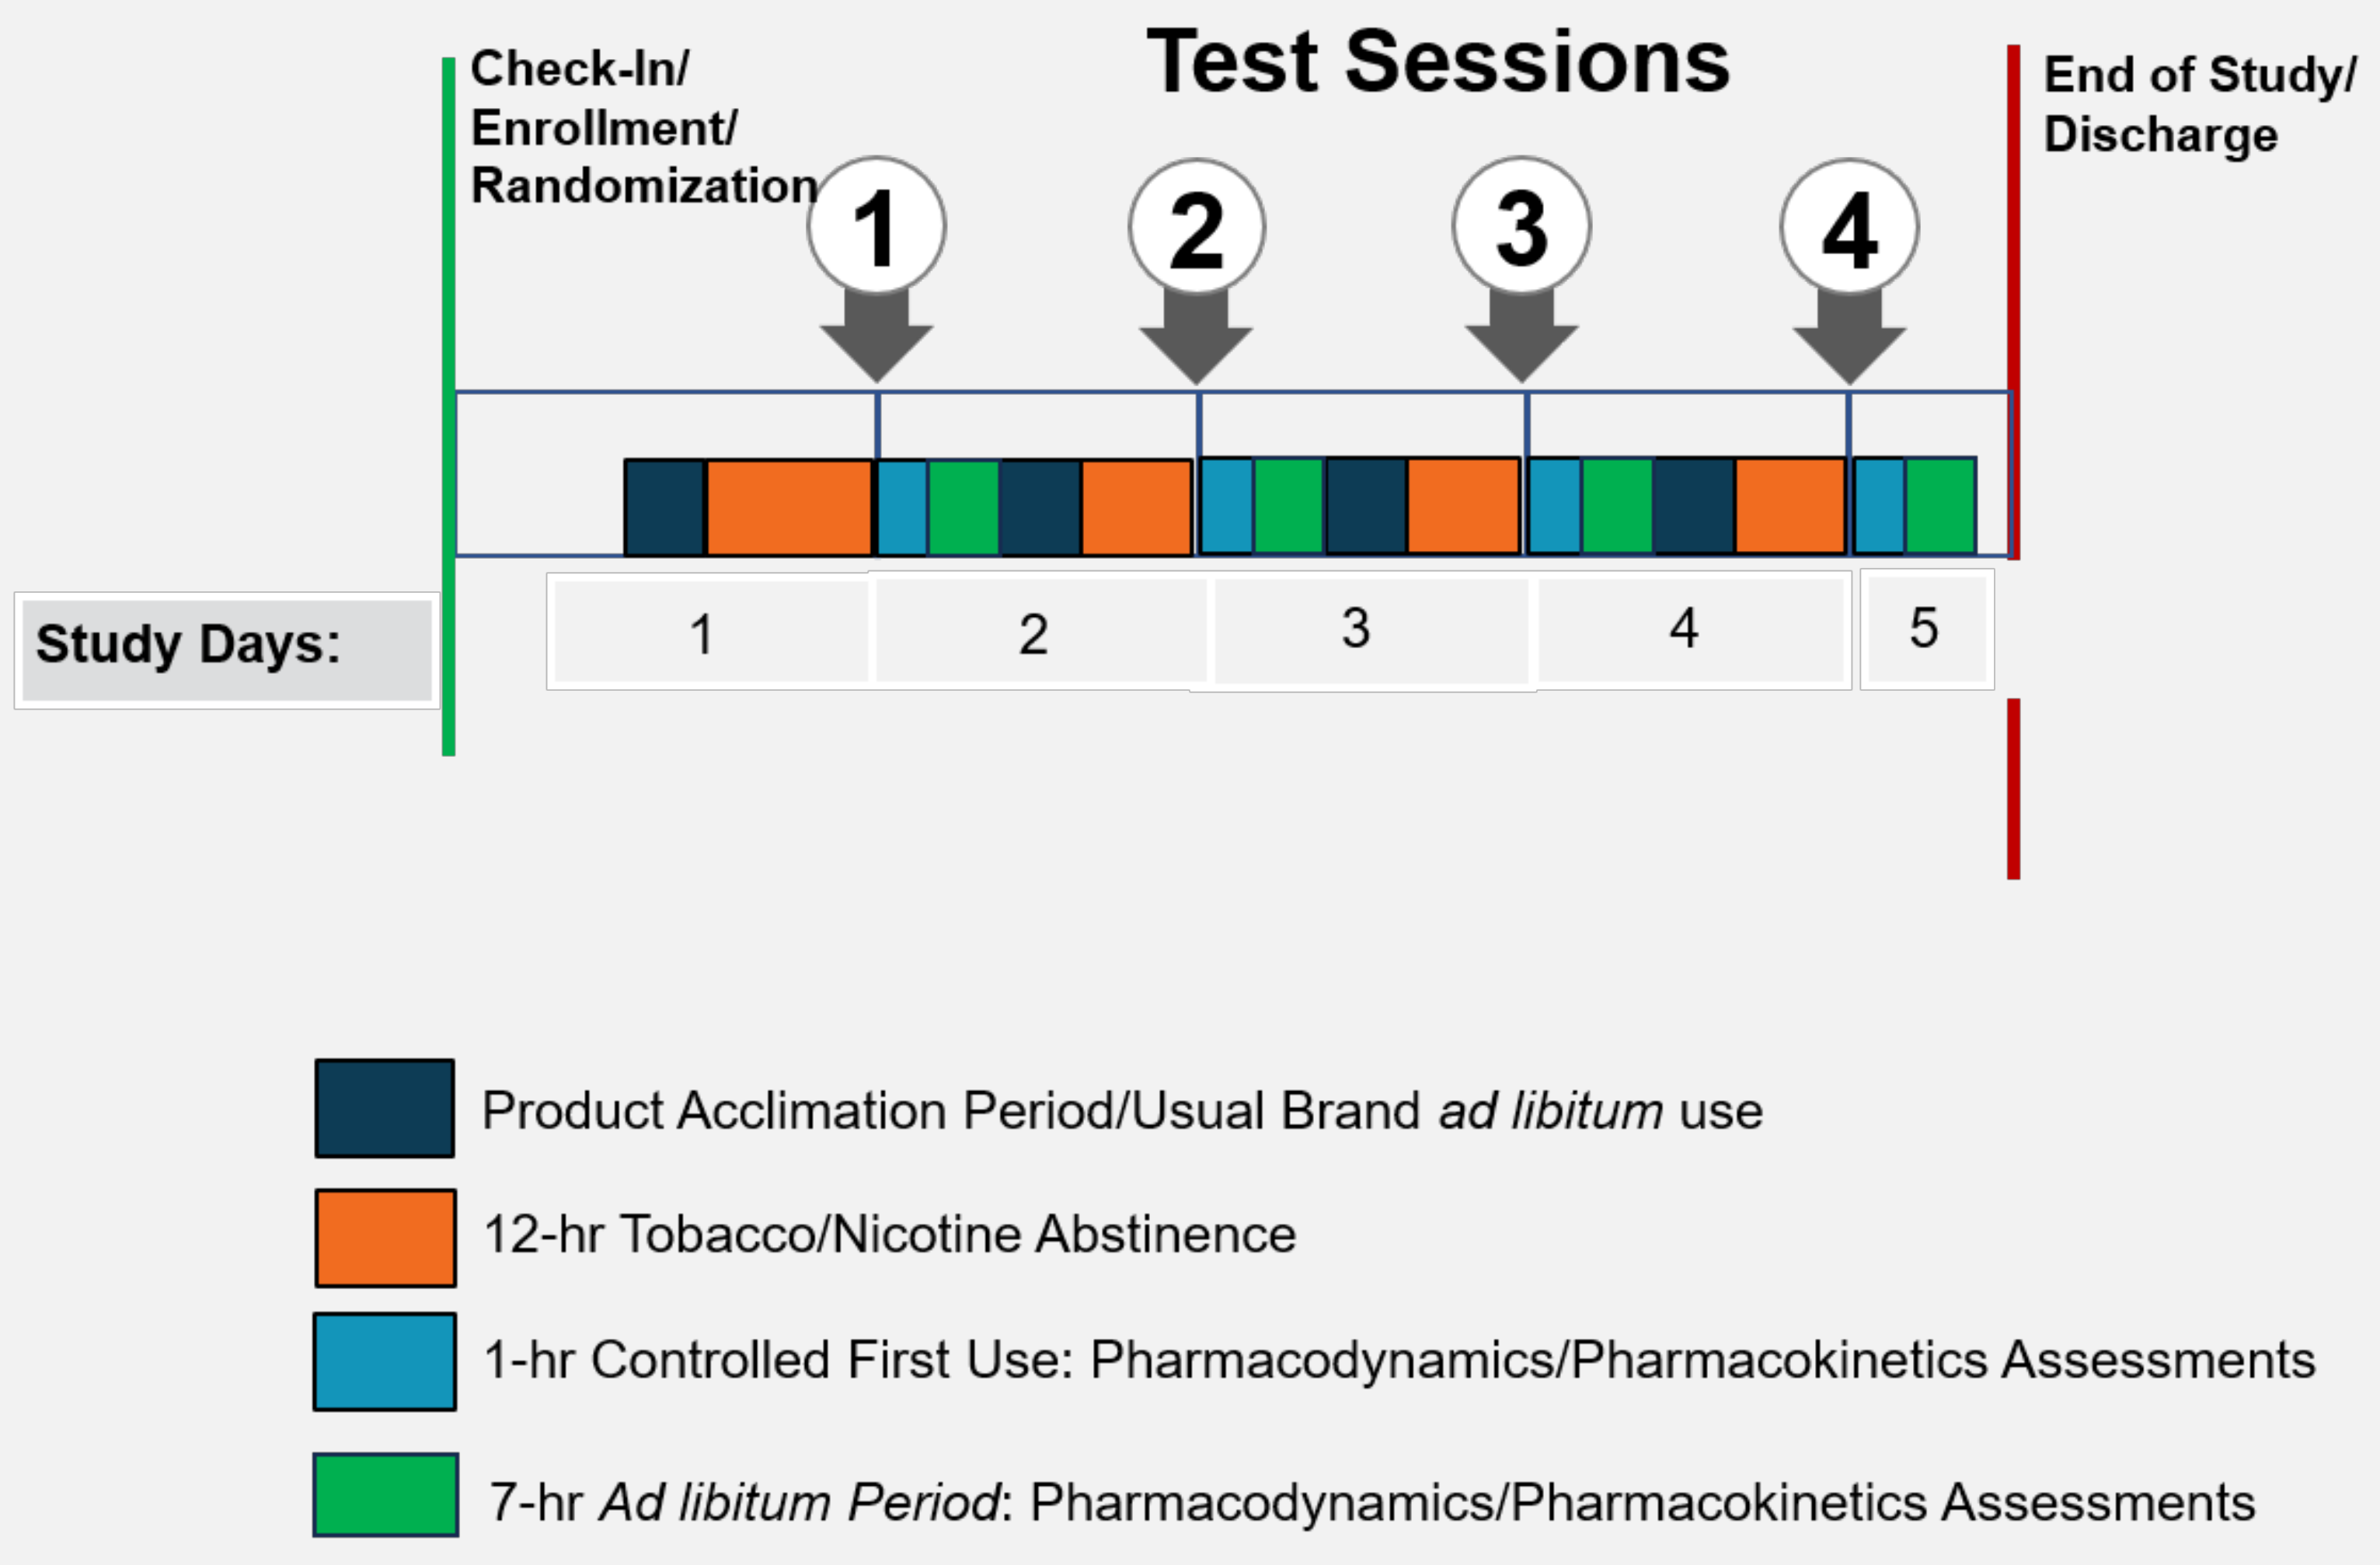
Test Sessions
Check-In/ Enrollment/ Randomization
End of Study/ Discharge
1
2
3
4
Study Days:
1
2
3
4
5
Product Acclimation Period/Usual Brand *ad libitum* use
12-hr Tobacco/Nicotine Abstinence
1-hr Controlled First Use: Pharmacodynamics/Pharmacokinetics Assessments
7-hr *Ad libitum Period*: Pharmacodynamics/Pharmacokinetics Assessments

Current Study Products and Comparators

High-AL Comparator IP

- UB filtered, non-menthol or menthol combustible cigarette

Velo PLUS MO IPs

- Velo PLUS Wintergreen-A 9 mg Nicotine
- Velo PLUS Wintergreen-B 9 mg Nicotine

Market Moist Snuff IP

- UB loose or pouched moist snuff

References

Nighbor T, Meredith S, Salazar E, Cartagena C, Koszowski B, Pickworth WB, Hull LC. A comparison of use topography and nicotine pharmacokinetics among loose and portioned smokeless tobacco users. Nicotine Tob Res. 2023 Apr;25(4):624-630. doi:10.1093/ntr/ntac195

Kanobe MN, Powell CY, Patrudu M, Baxter SA, Tapia MA, Darnell J, Prevette K, Gibson AG, Ayoku SA, Campbell L, Coffield JW, Keyser BM, Ganesh BS, Gale N, Jordan KG. Randomized crossover clinical studies to assess abuse liability and nicotine pharmacokinetics of Velo Oral Nicotine pouches. Front Pharmacol. 2025;16:1547073. doi:10.3389/fphar.2025.1547073

U.S. Food and Drug Administration, Center for Tobacco Products (CTP). Roundtable on Premarket Tobacco Product Application (PMTA) Submissions for Electronic Nicotine Delivery Systems (ENDS) Products. February 10, 2026, Silver Spring, MD. FDA. Available at: FDA PMTA Roundtable Event Page.

Evolution of Study Designs

Cigarette/NRT Comparator Design

- 30 minute controlled product use
- PK and subjective effect assessment through 240 min
- High AL comparator – UBCC
- Low AL comparator – NRT

✓ ***High & low AL comparators, acute PK profile and product liking***

✗ ***No information regarding nicotine uptake with continued product use behavior***

Moist Snuff Comparator Design

- 30 minute controlled product use
- PK and subjective effect assessment through 240 min
- Comparator – Usual Brand Moist Snuff
- Low AL comparator – none

✓ ***Oral product comparator, PK profile and product liking***

✗ ***No information regarding nicotine uptake with continued product use behavior***

Conclusion

This evolution of AL study design for modern oral nicotine pouches reflects a broader understanding that assessments focused primarily on acute subjective effects and nicotine PK represent only part of the overall AL profile of a product. Integrating acute PK and subjective assessments with extended ad libitum use evaluation while including combustible and moist snuff comparators provides a more comprehensive framework for characterizing modern oral nicotine pouches and may provide regulatory-relevant insights of nicotine uptake and product use behavior beyond single-use assessments.

- Previous single use PK studies remain valuable for characterizing acute nicotine uptake and product appeal.
- Acute assessments alone provide limited information about continued product use behavior.
- A 7-hour *ad libitum* phase characterizes repeated use and sustained exposure.
- Integrating combustible cigarette and moist snuff comparators provides regulatory relevant AL context.

REYNOLDS
A BETTER TOMORROW

Contacts
IKristen Prevette: prevetk1@rjrt.com

Point your phone's camera at the QR code to find our library of publications

Follow us:

www.ReynoldsScience.com

@RAI_News